RenMab Mouse: A Leading Platform for Fully Human Antibody Generation

Chaoshe Guo[1], Benny Yang[2], Huizhen Zhao[2], Yabo Zhang[2], Lili Liu[2], Hui Lu[2], Shuwen Huang[2], Yuelei Shen[2]

[1]Biocytogen Boston Corp., Wakefield, MA, USA; [2]Beijing Biocytogen Co., Ltd., Beijing, China

ABSTRACT

With the development of immuno-oncology, therapeutic antibodies have been proven to be extraordinarily effective for cancer treatment. Conventional human antibody discovery process can be divided into stages including target selection and validation, screening preparation, hits generation, leads selection and optimization, and clinical candidate selection. To accelerate antibody development process, Biocytogen has developed RenMab™ Mouse, a fully human antibody mouse whose entire mouse variable regions were replaced by human Immunoglobulin heavy chain and κ light chain through Biocytogen's unique Mb-scale chromosome engineering technology. RenMab™ Mouse provides an efficient therapeutic antibody discovery platform for fully human antibody hit generation and characterization. We have characterized RenMab™ Mouse with the following features.

1. The entire mouse variable regions were replaced with complete human genome DNA in situ for complete human & mouse regulatory elements. The gene regulation of RenMab™ mouse is highly consistent with that of human.
2. Mouse constant region remains to ensure proper B cell development and maturation. Immune system of RenMab™ Mouse has been proven to be almost identical to that of wild type mouse. RenMab™ Mouse showed normal antibody immune responses to antigens.
3. RenMab™ Mouse generates a highly diverse repertoire of fully human antibody variable regions through V(D)J recombination. This capability can lead to promising hits for downstream leads and candidate selection in human therapeutic antibody discovery programs.

RESULTS AND DISCUSSIONS

Schematic of Humanization in The RenMab™ Mouse

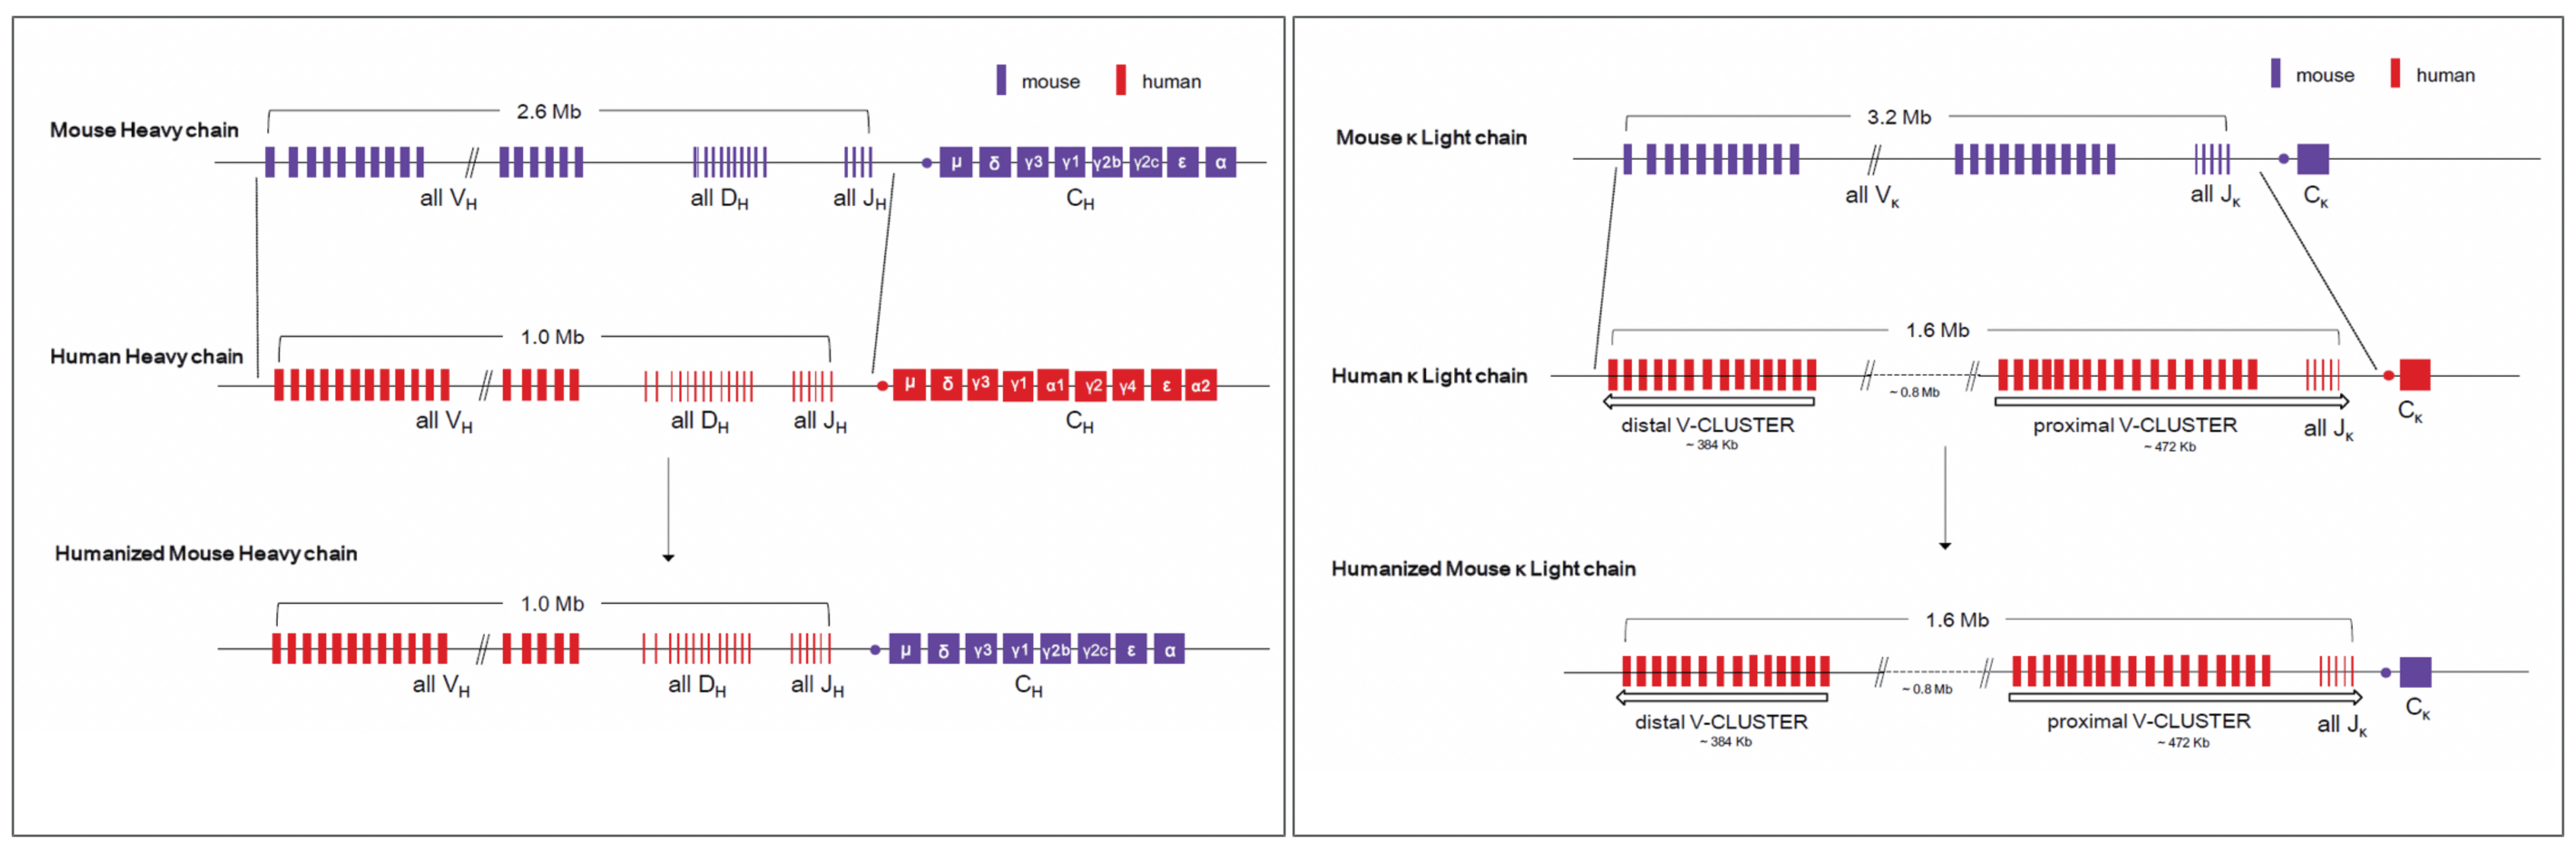
Human
Mouse
Mouse Heavy chain
2.6 Mb
Human Heavy chain
1.0 Mb
Humanized Mouse Heavy chain
Mouse κ Light chain
3.2 Mb
Human κ Light chain
1.6 Mb
distal V-CLUSTER
proximal V-CLUSTER
Humanized Mouse κ Light chain

Whole mouse variable regions of the heavy and k light chains are replaced by full human heavy chain VDJ segment and light chain VJ loci *in situ*

I. RenMab™ Mouse demonstrates a similar immune system to that of WT mouse

RenMab Mice immune cells profiling suggest a comparable immune system with wild type mice

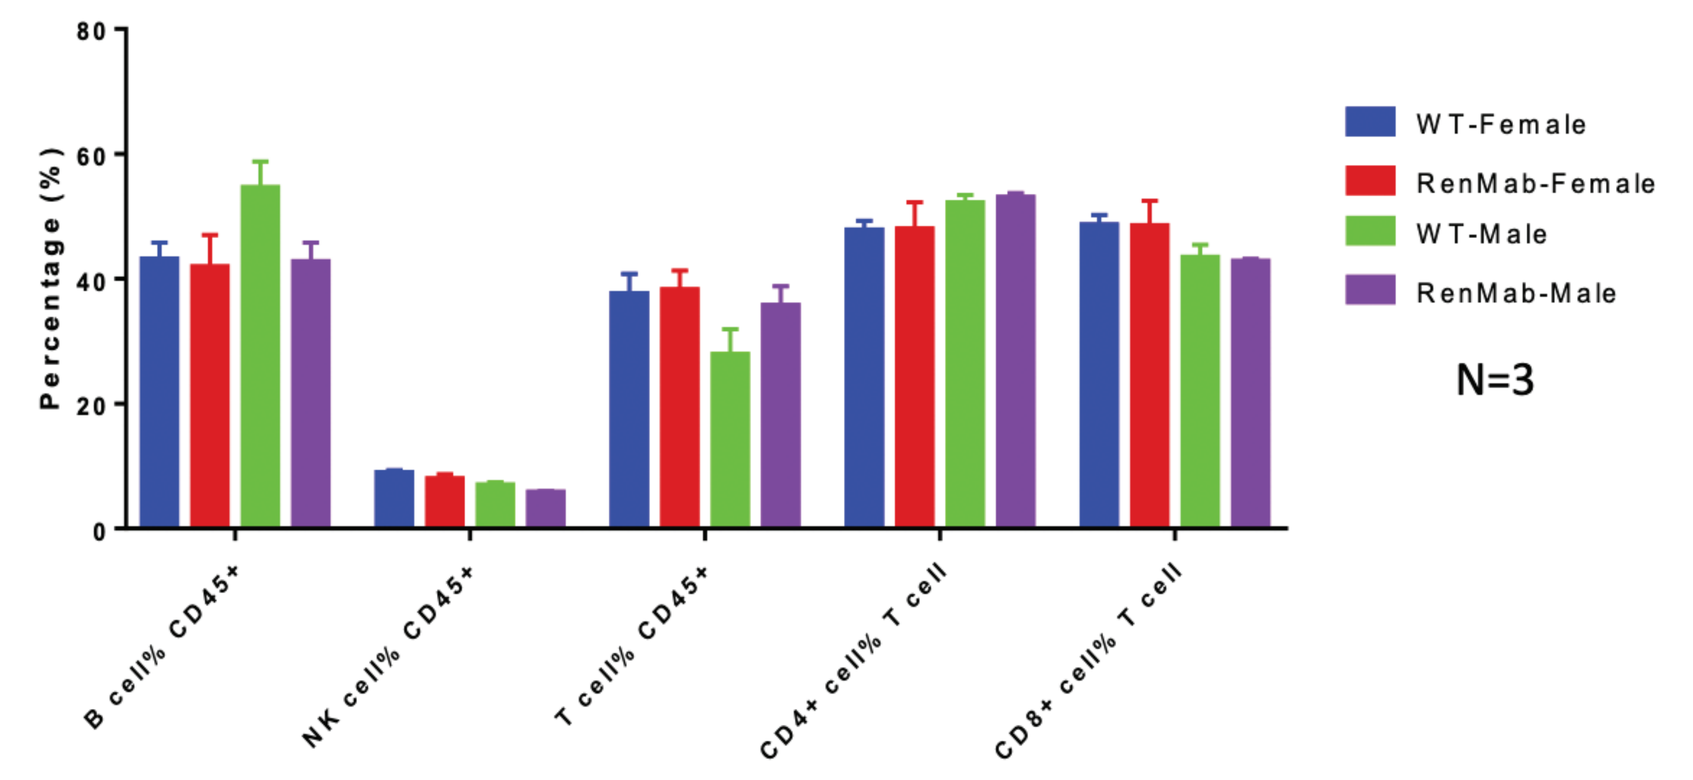
Percentage (%)
B cell% CD45+
NK cell% CD45+
T cell% CD45+
CD4+ cell% T cell
CD8+ cell% T cell
WT-Female
RenMab-Female
WT-Male
RenMab-Male
N=3

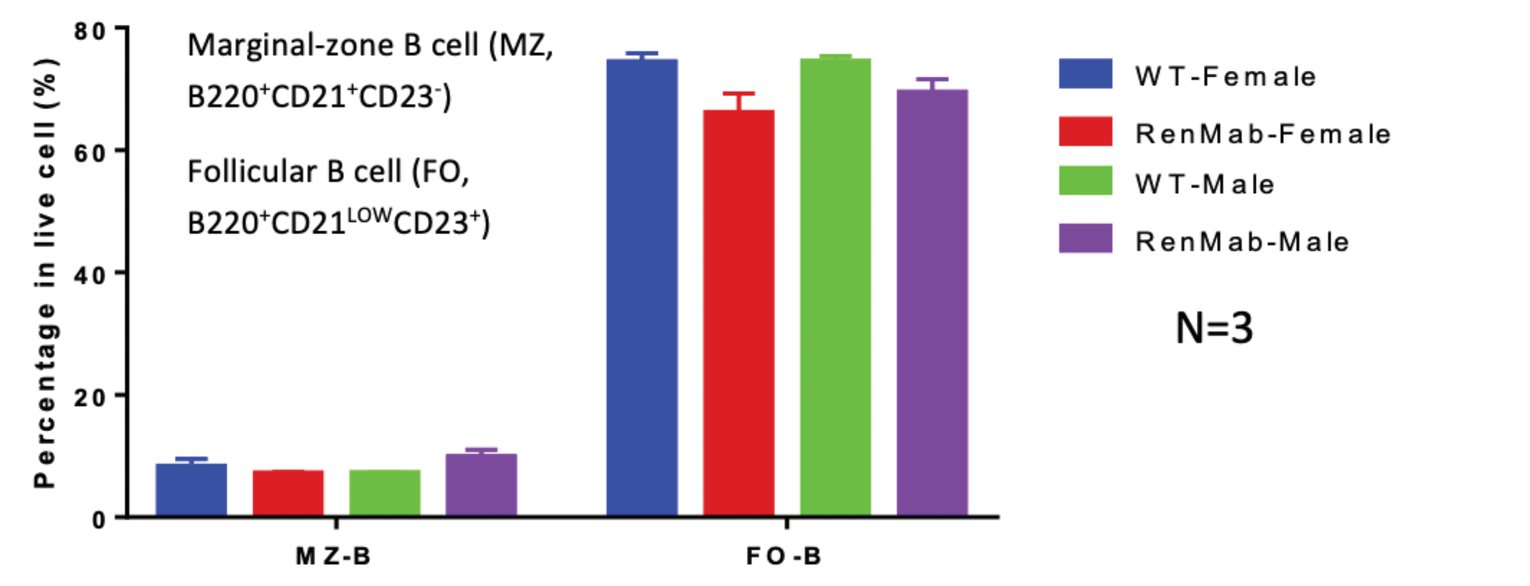
Marginal-zone B cell (MZ, B220+CD21+CD23-)
Follicular B cell (FO, B220+CD21LOWCD23+)
Percentage in live cell (%)
MZ-B
FO-B
WT-Female
RenMab-Female
WT-Male
RenMab-Male
N=3

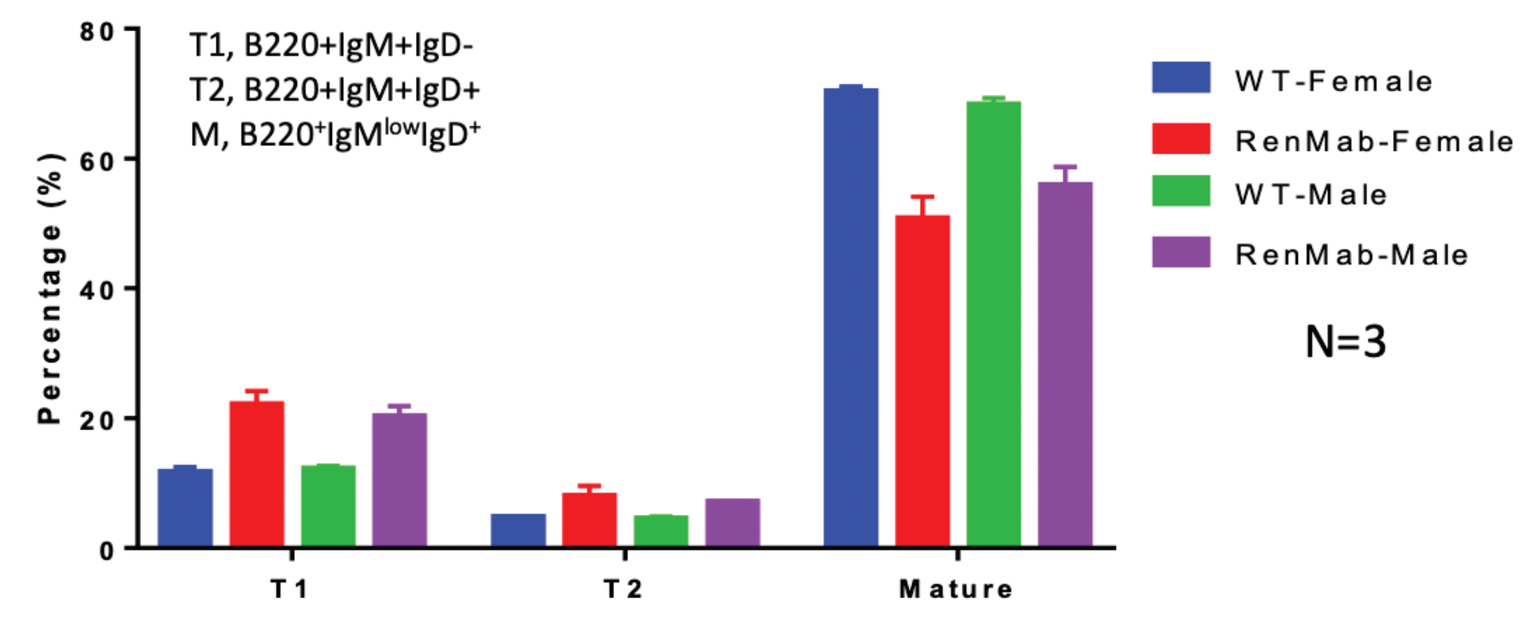
T1, B220+IgM+IgD-
T2, B220+IgM+IgD+
M, B220+IgMlowIgD+
Percentage (%)
T1
T2
Mature
WT-Female
RenMab-Female
WT-Male
RenMab-Male
N=3

• Immune cells profiling in spleen (left) and B cell development in spleen (right) were evaluated.
• No significant difference was observed between RenMab™ and wild type mice.
• In RenMab™ mice, slight delay in B cell development was observed.

II. Full human VDJ expression in the variable domains of heavy chain and kappa light chain genes

The RenMab™ mouse shows WT-like expression of antibody levels and full usage of human heavy chain V, D, and J domain segments. Similarly, mouse kappa chain segments were replaced with human V and J segments and also show WT-like expression of antibody levels.

RenMab™ Naive Mouse Heavy Chain IGHV Germline Usage

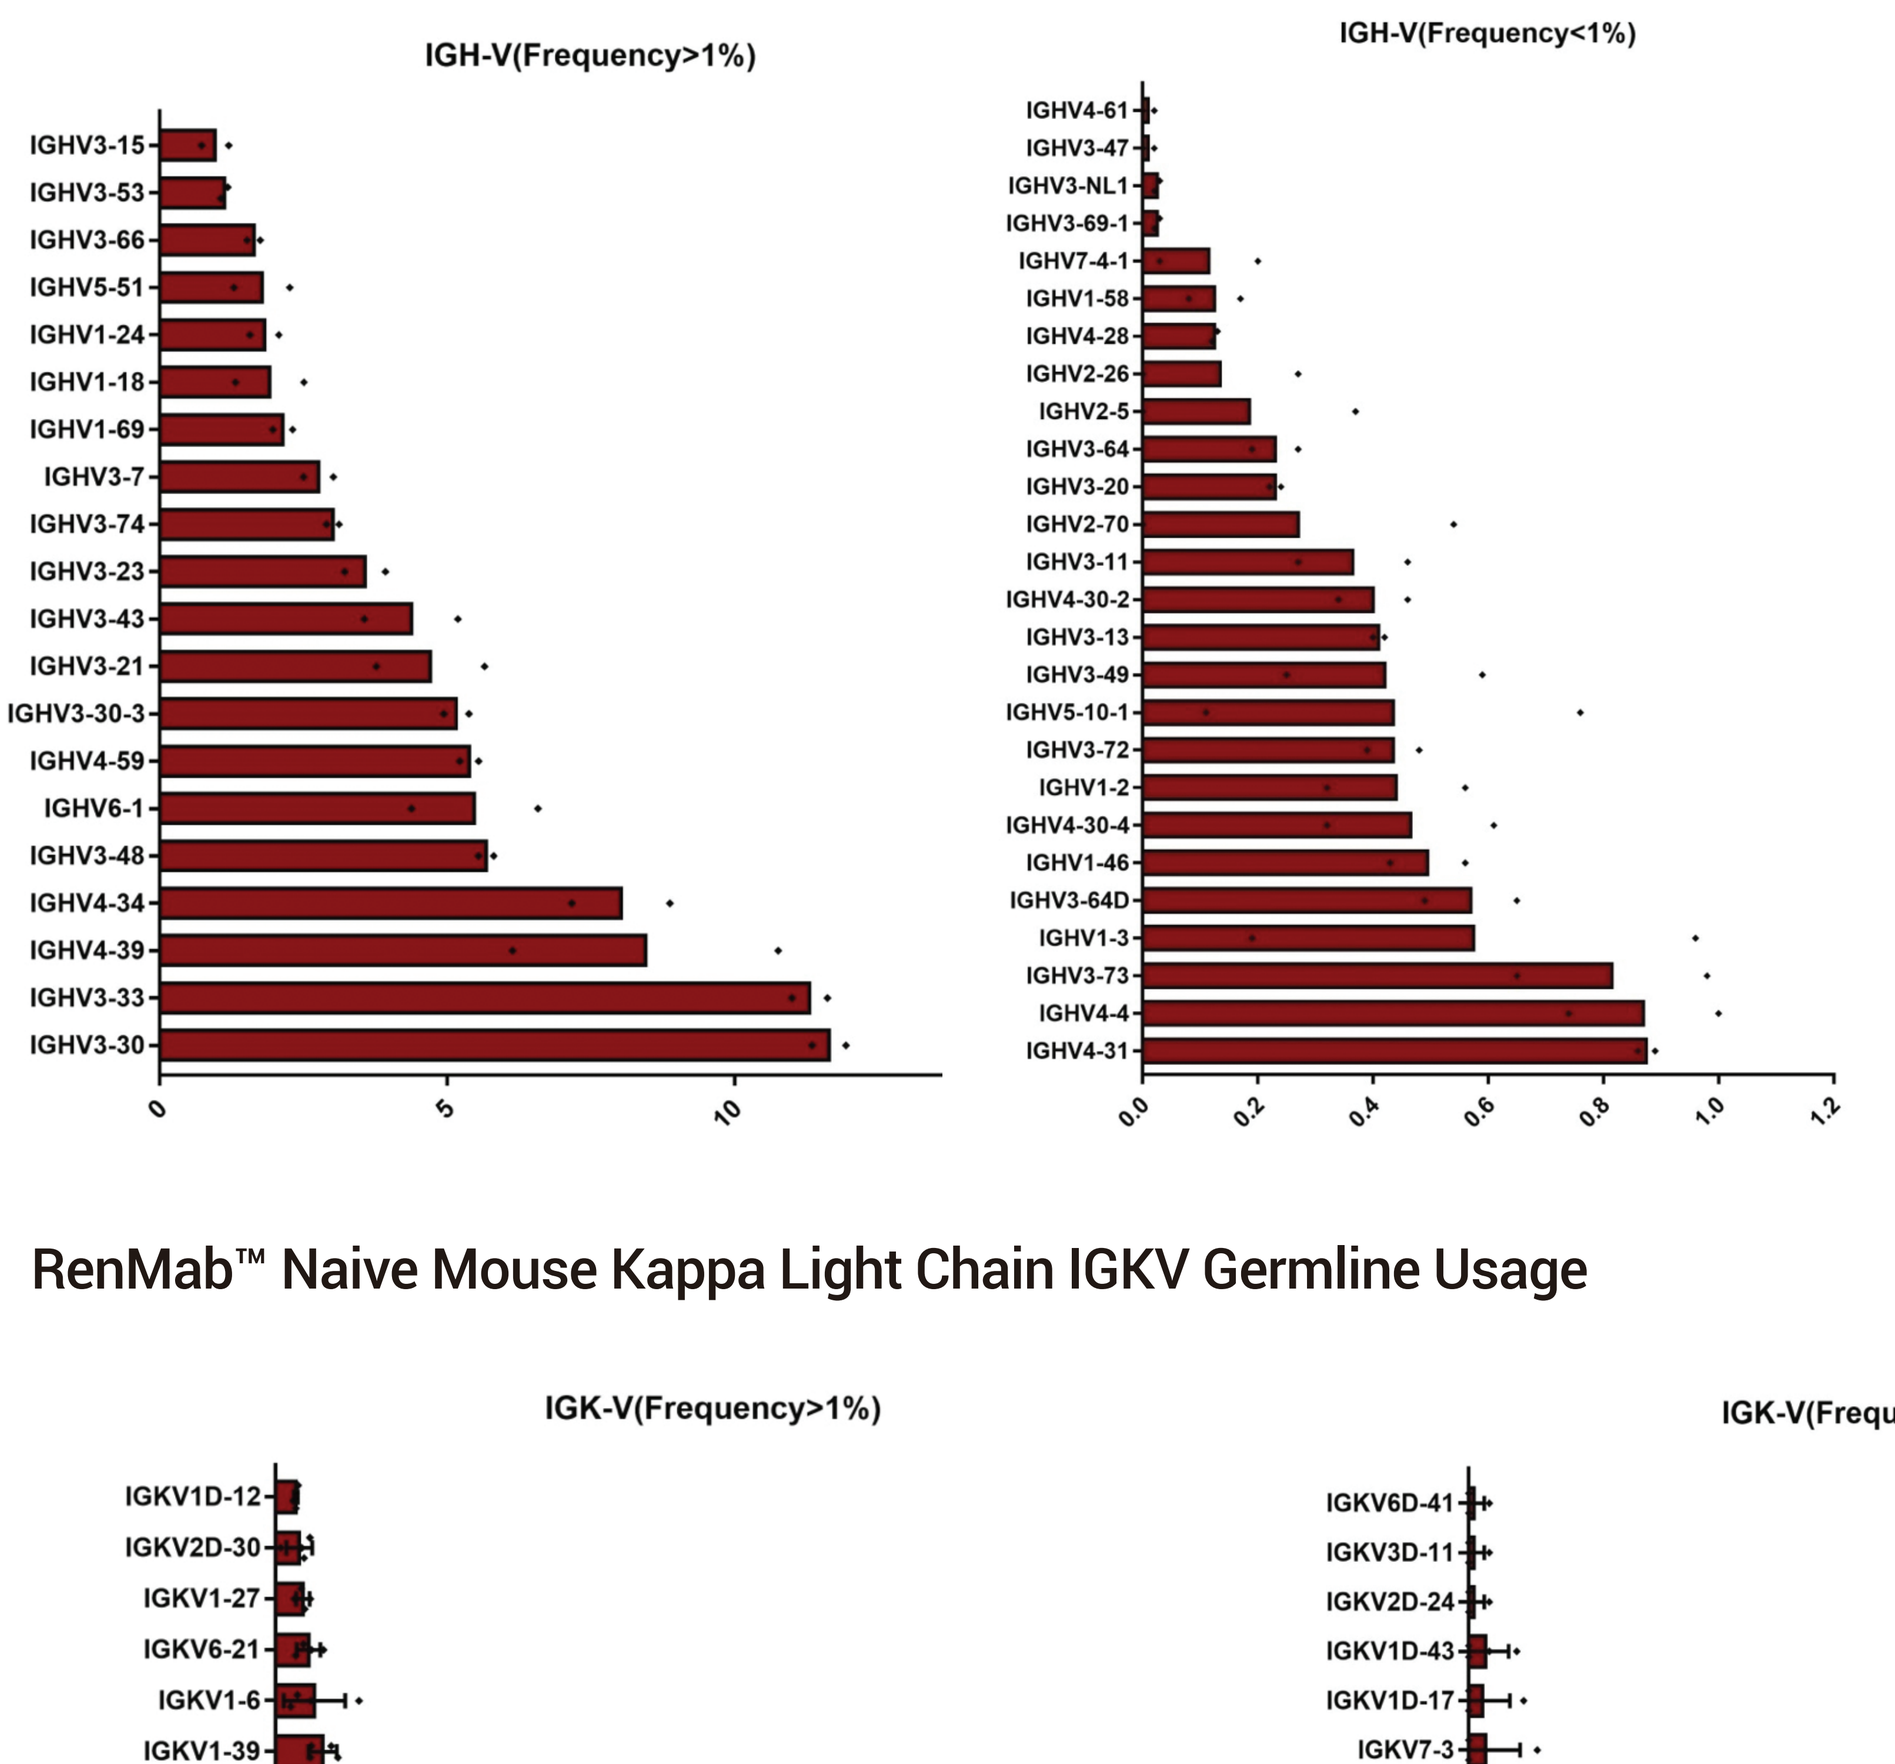
IGH-V(Frequency>1%)
IGH-V(Frequency<1%)

RenMab™ Naive Mouse Heavy Chain IGHD & IGHJ Germline Usage

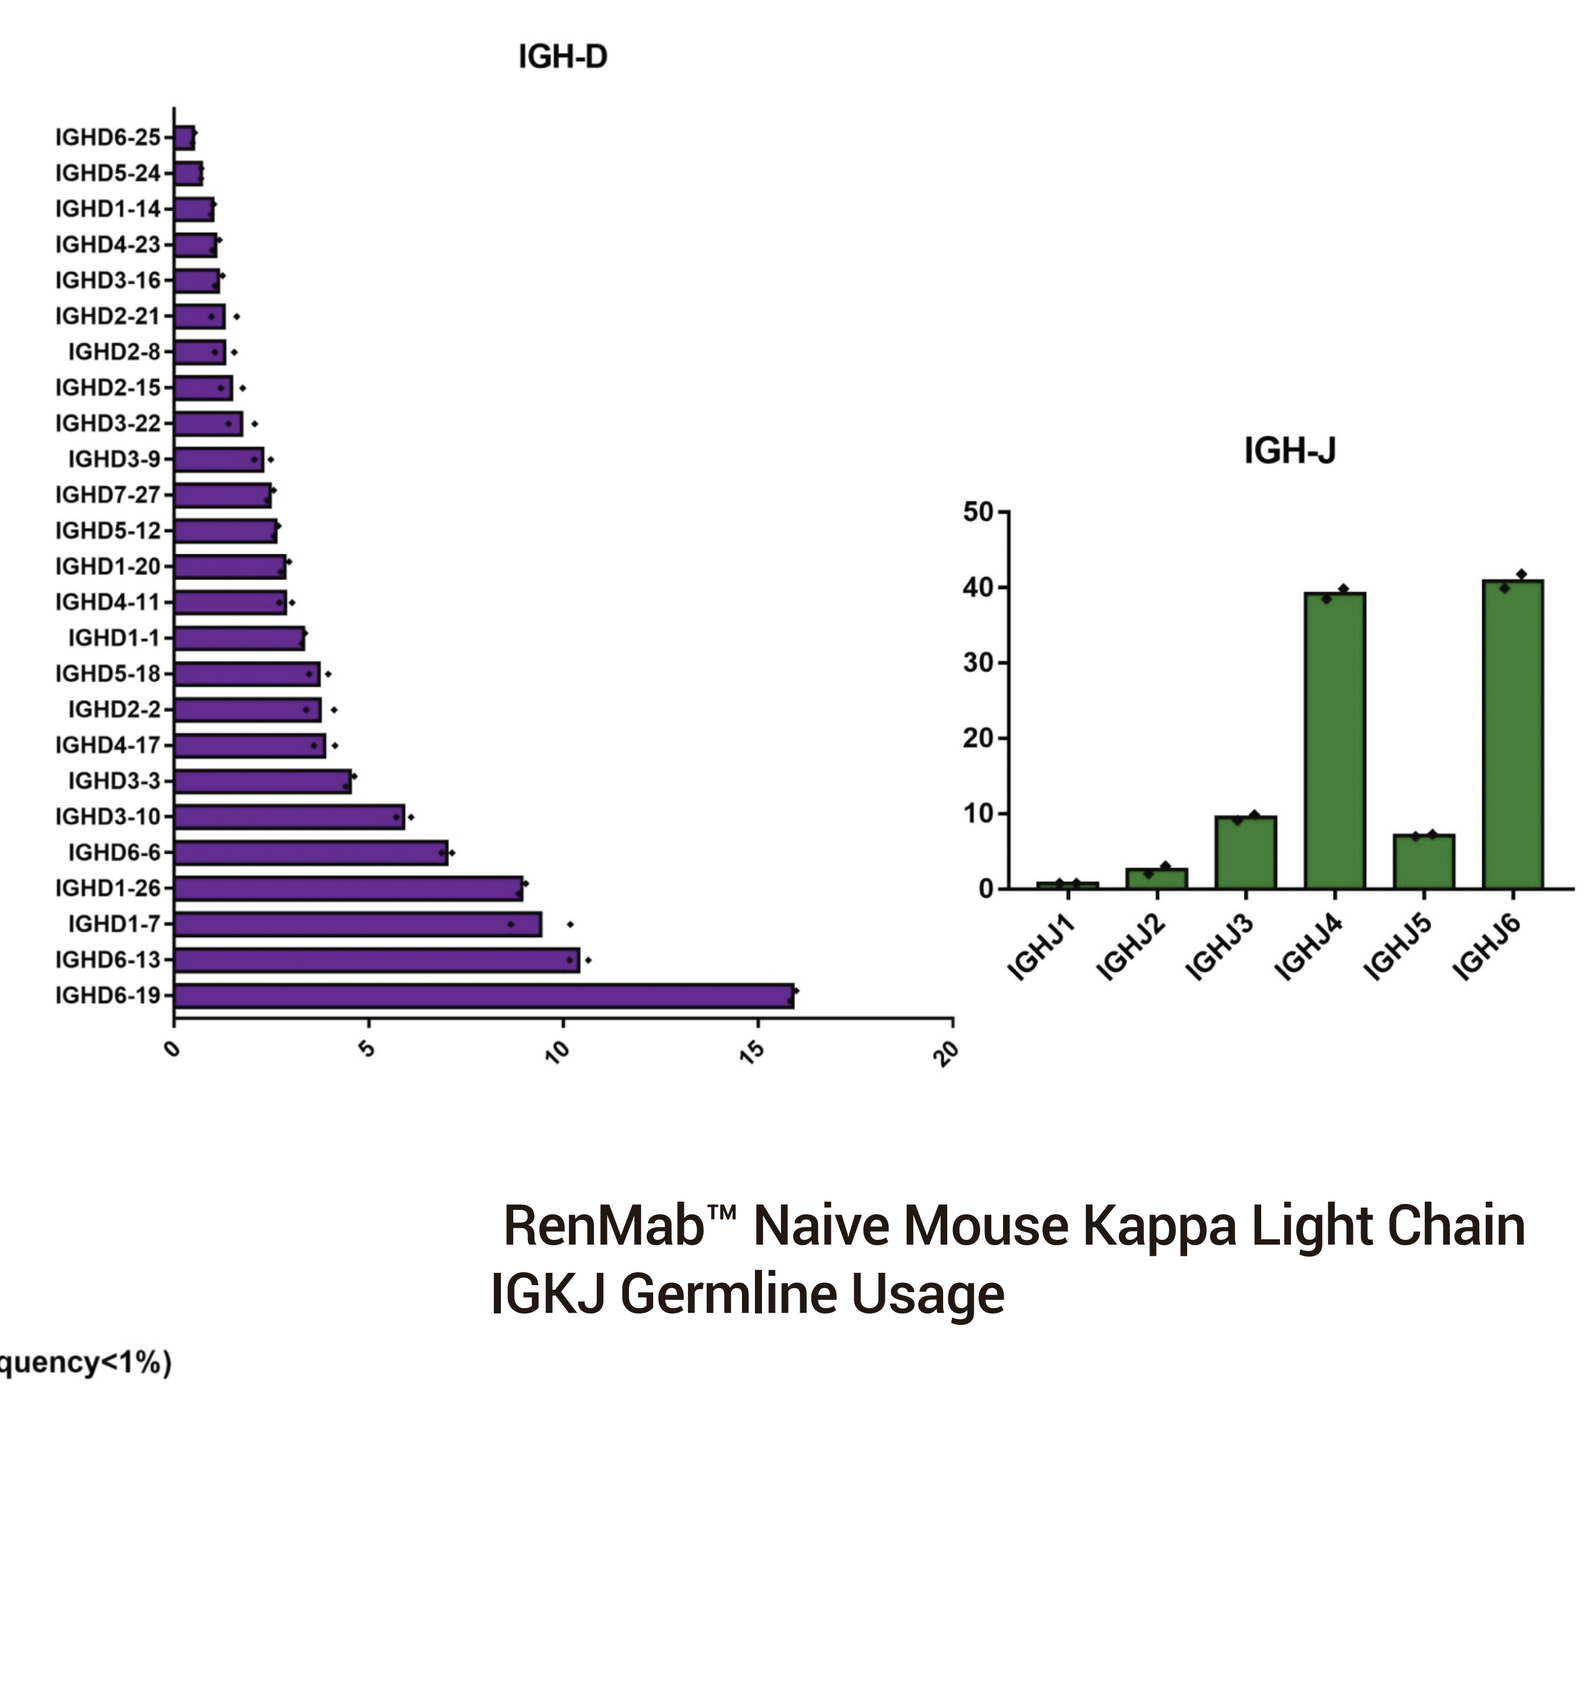
IGH-D
IGH-J

RenMab™ Naive Mouse Kappa Light Chain IGKV Germline Usage

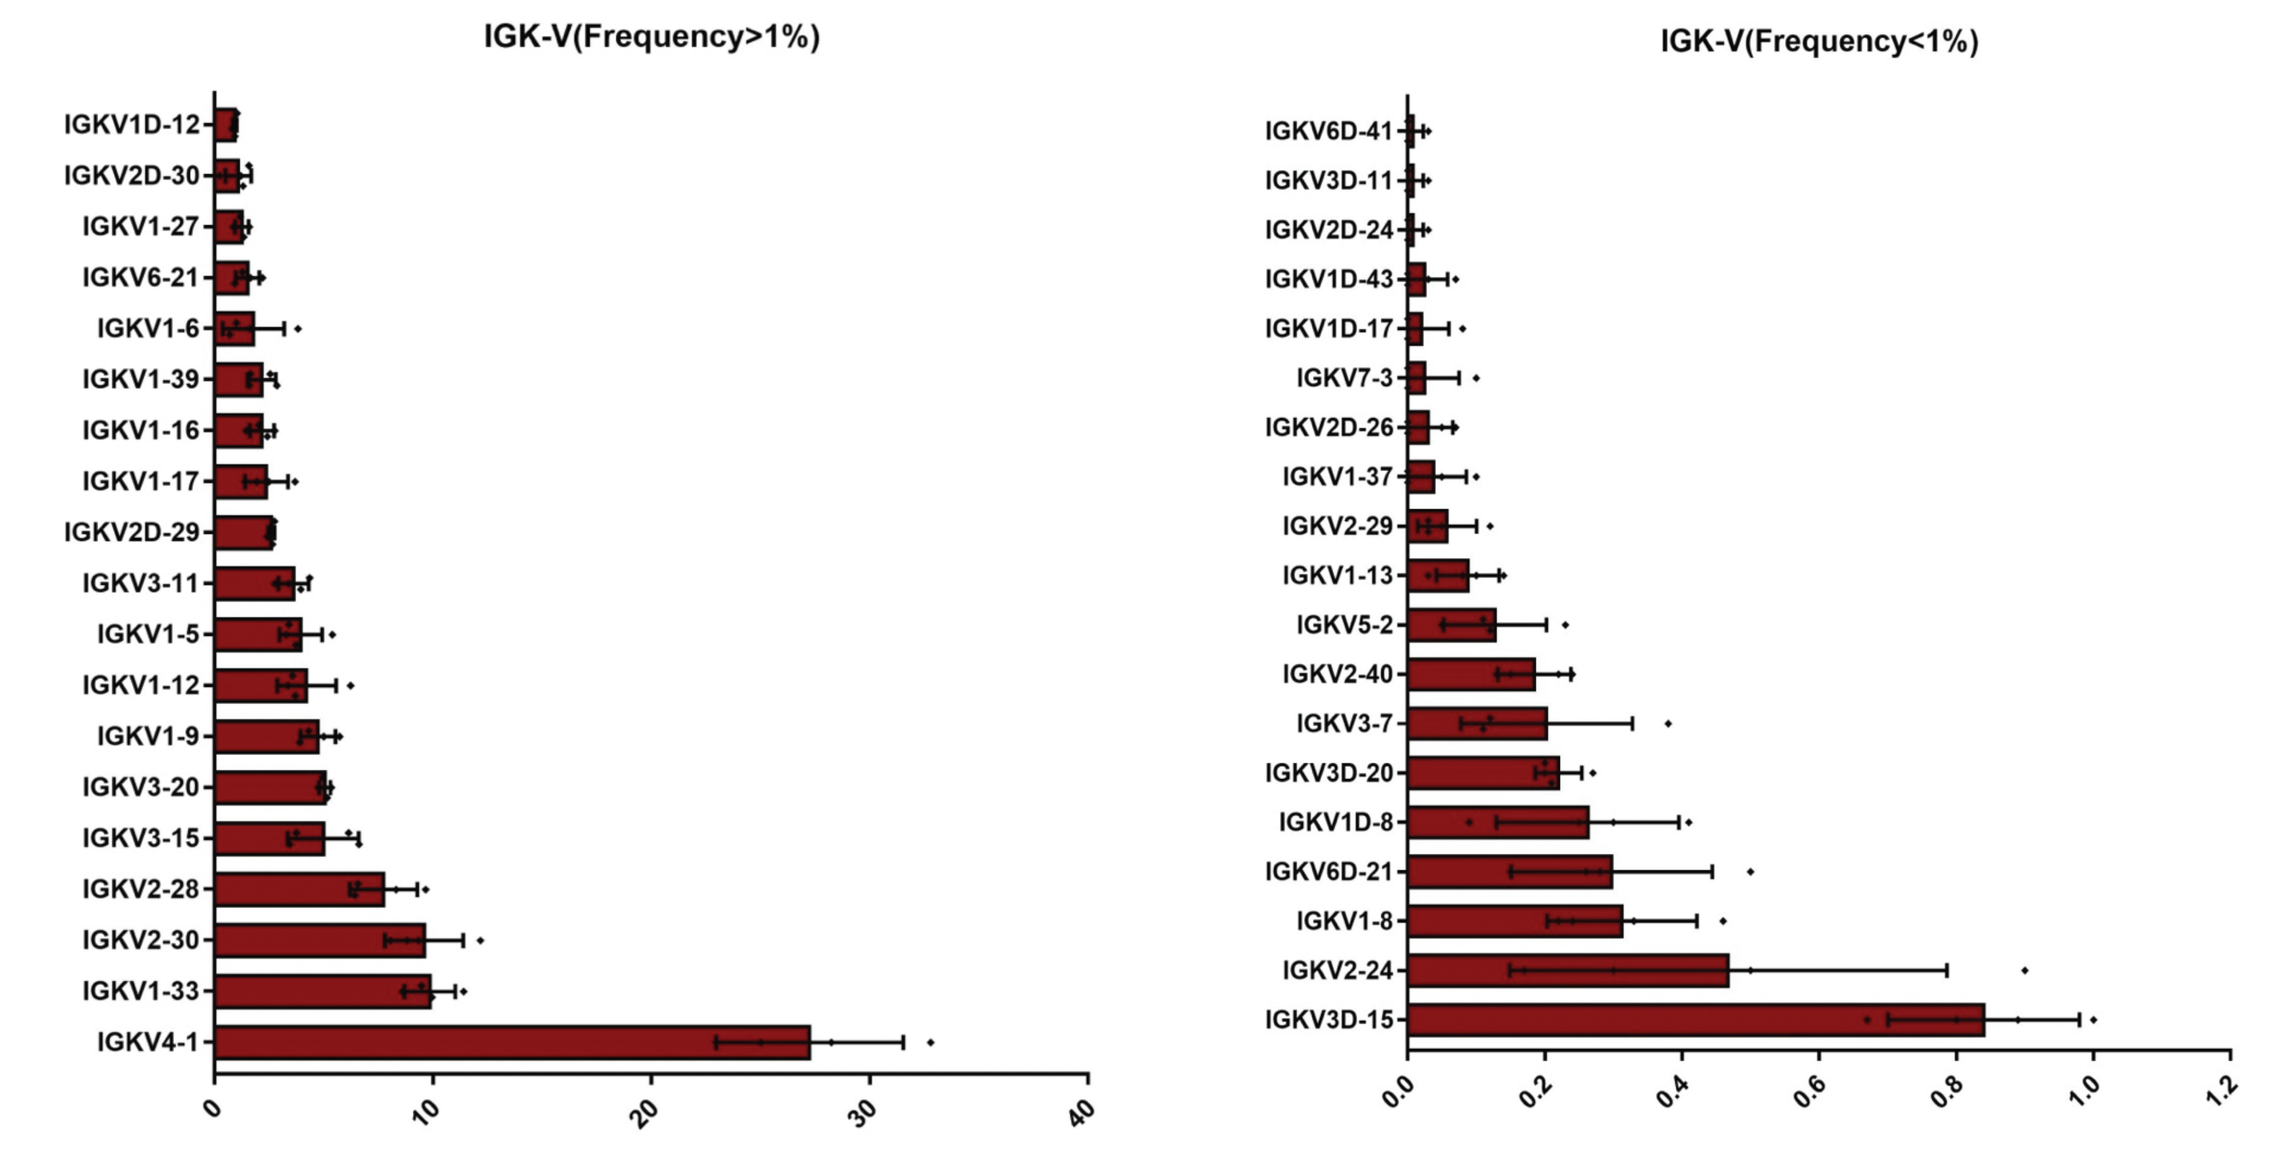
IGK-V(Frequency>1%)
IGK-V(Frequency<1%)

RenMab™ Naive Mouse Kappa Light Chain IGKJ Germline Usage

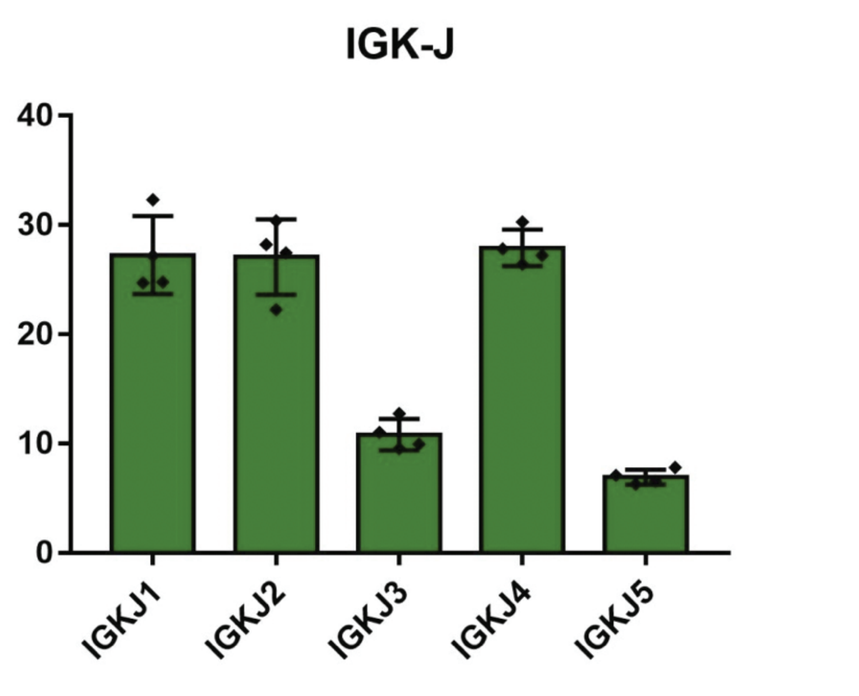
IGK-J
IGKJ1
IGKJ2
IGKJ3
IGKJ4
IGKJ5

III. RenMab™ Mouse produces human-like antibody repertoire as shown by CDR3 analysis

CDRH3 length (amino acids) IgM-Native RenMab™ spleen cell (n=2)

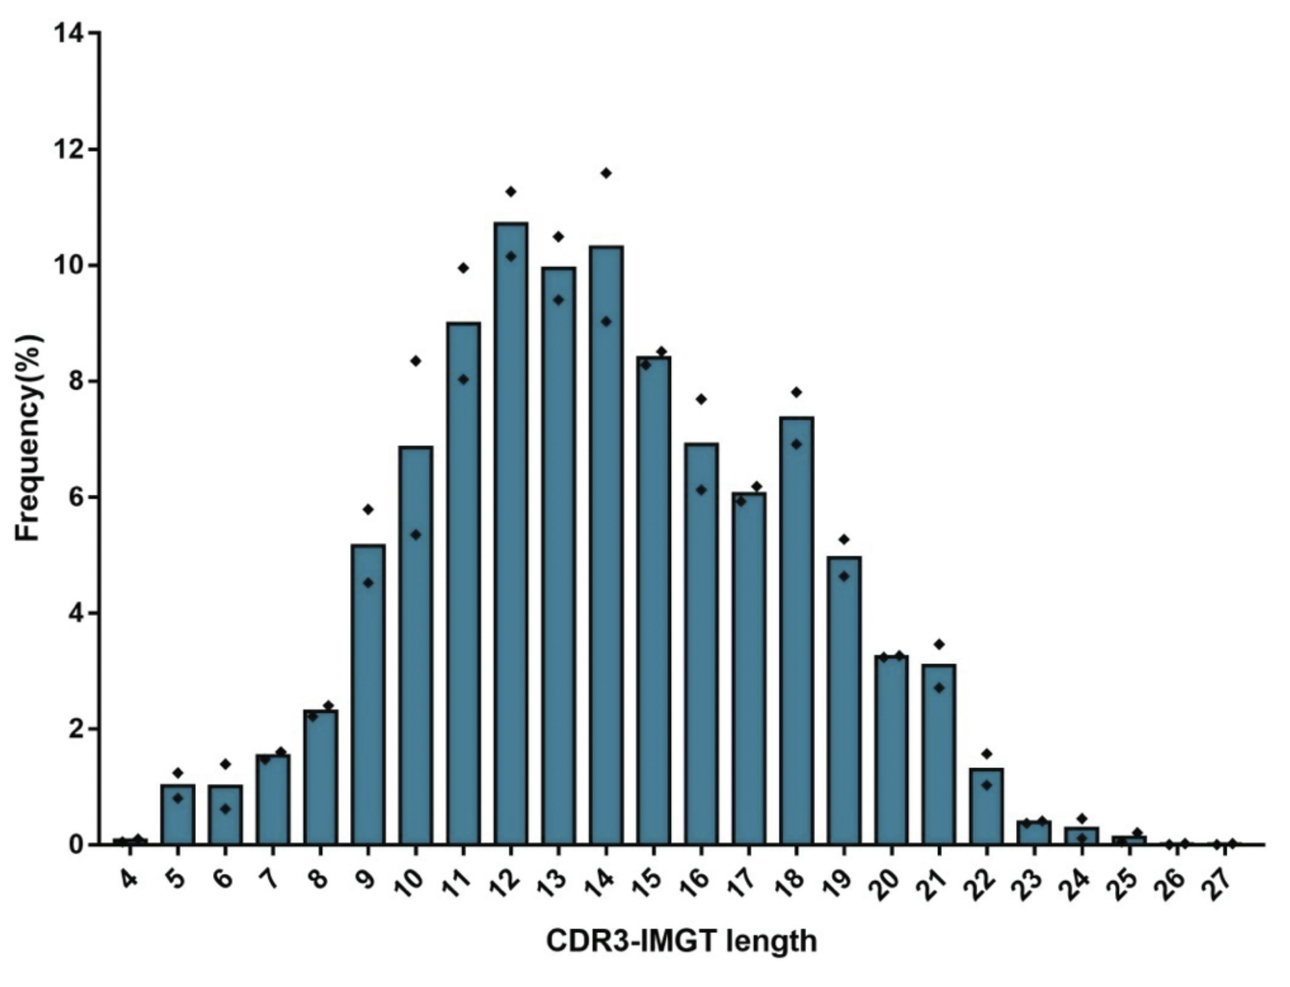
Frequency(%)
CDR3-IMGT length

Heavy chain CDR3 length distribution: median length 14.2 amino acids

RenMab™ Heavy Chain CDR3 Amino Acid Usage Frequency

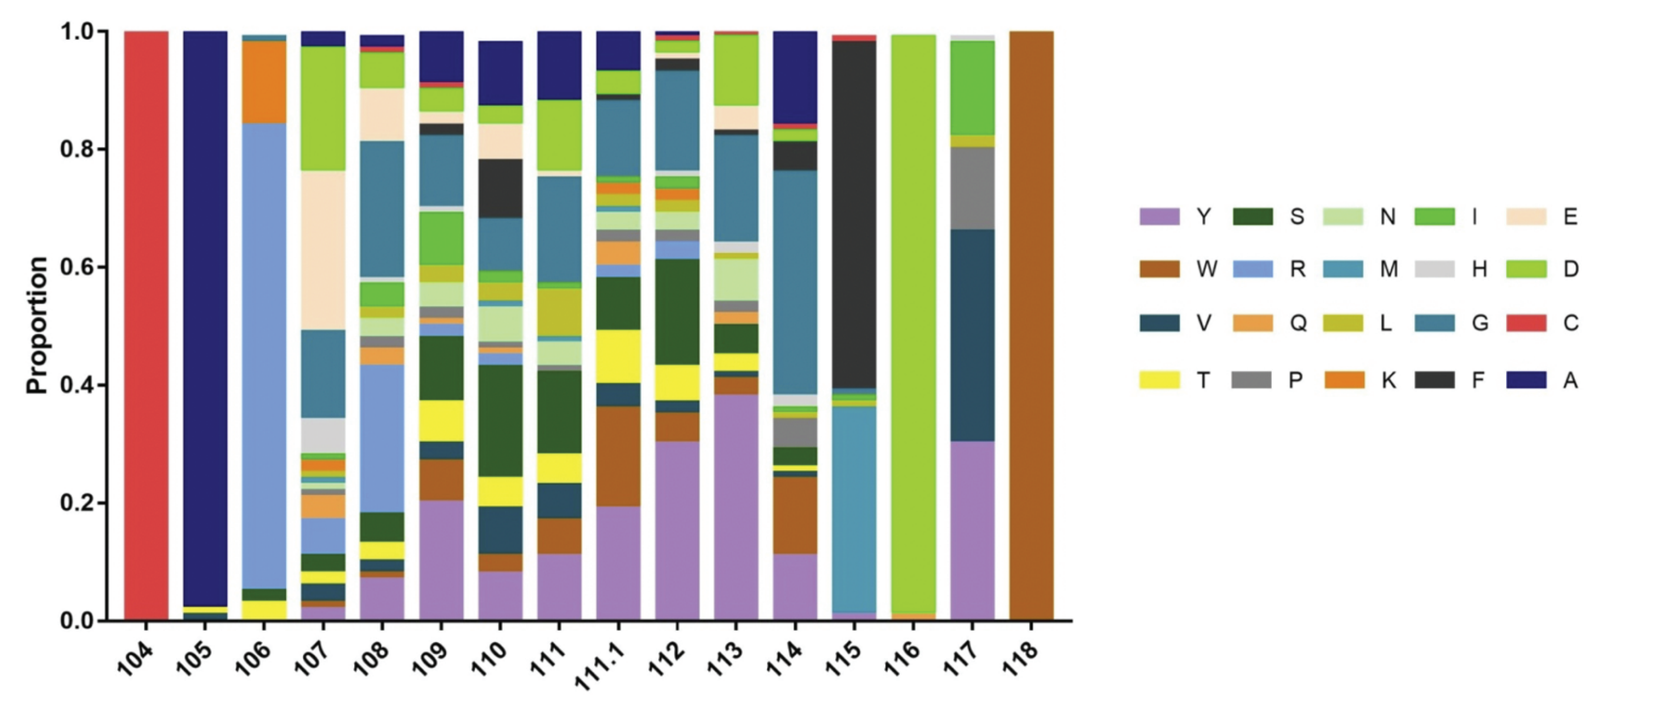
Proportion

Human-like heavy chain V-D-J recombination pattern: (1) length-dependent usage of tyrosine; (2) preferred usage of D, E and G at IMGT position 107; (3) IMGT position 115 and 117 are conserved due to human JH4 and JH6 usage; (4) Usage of cysteine residue in CDR3, indicative of human DH2 gene family usage in V-D-J recombination (data not shown).

IV. RenMab™ Mouse demonstrates highly similar immune responses compared to WT mouse

Robust immune response in RenMab™ elicited by a panel of antigens

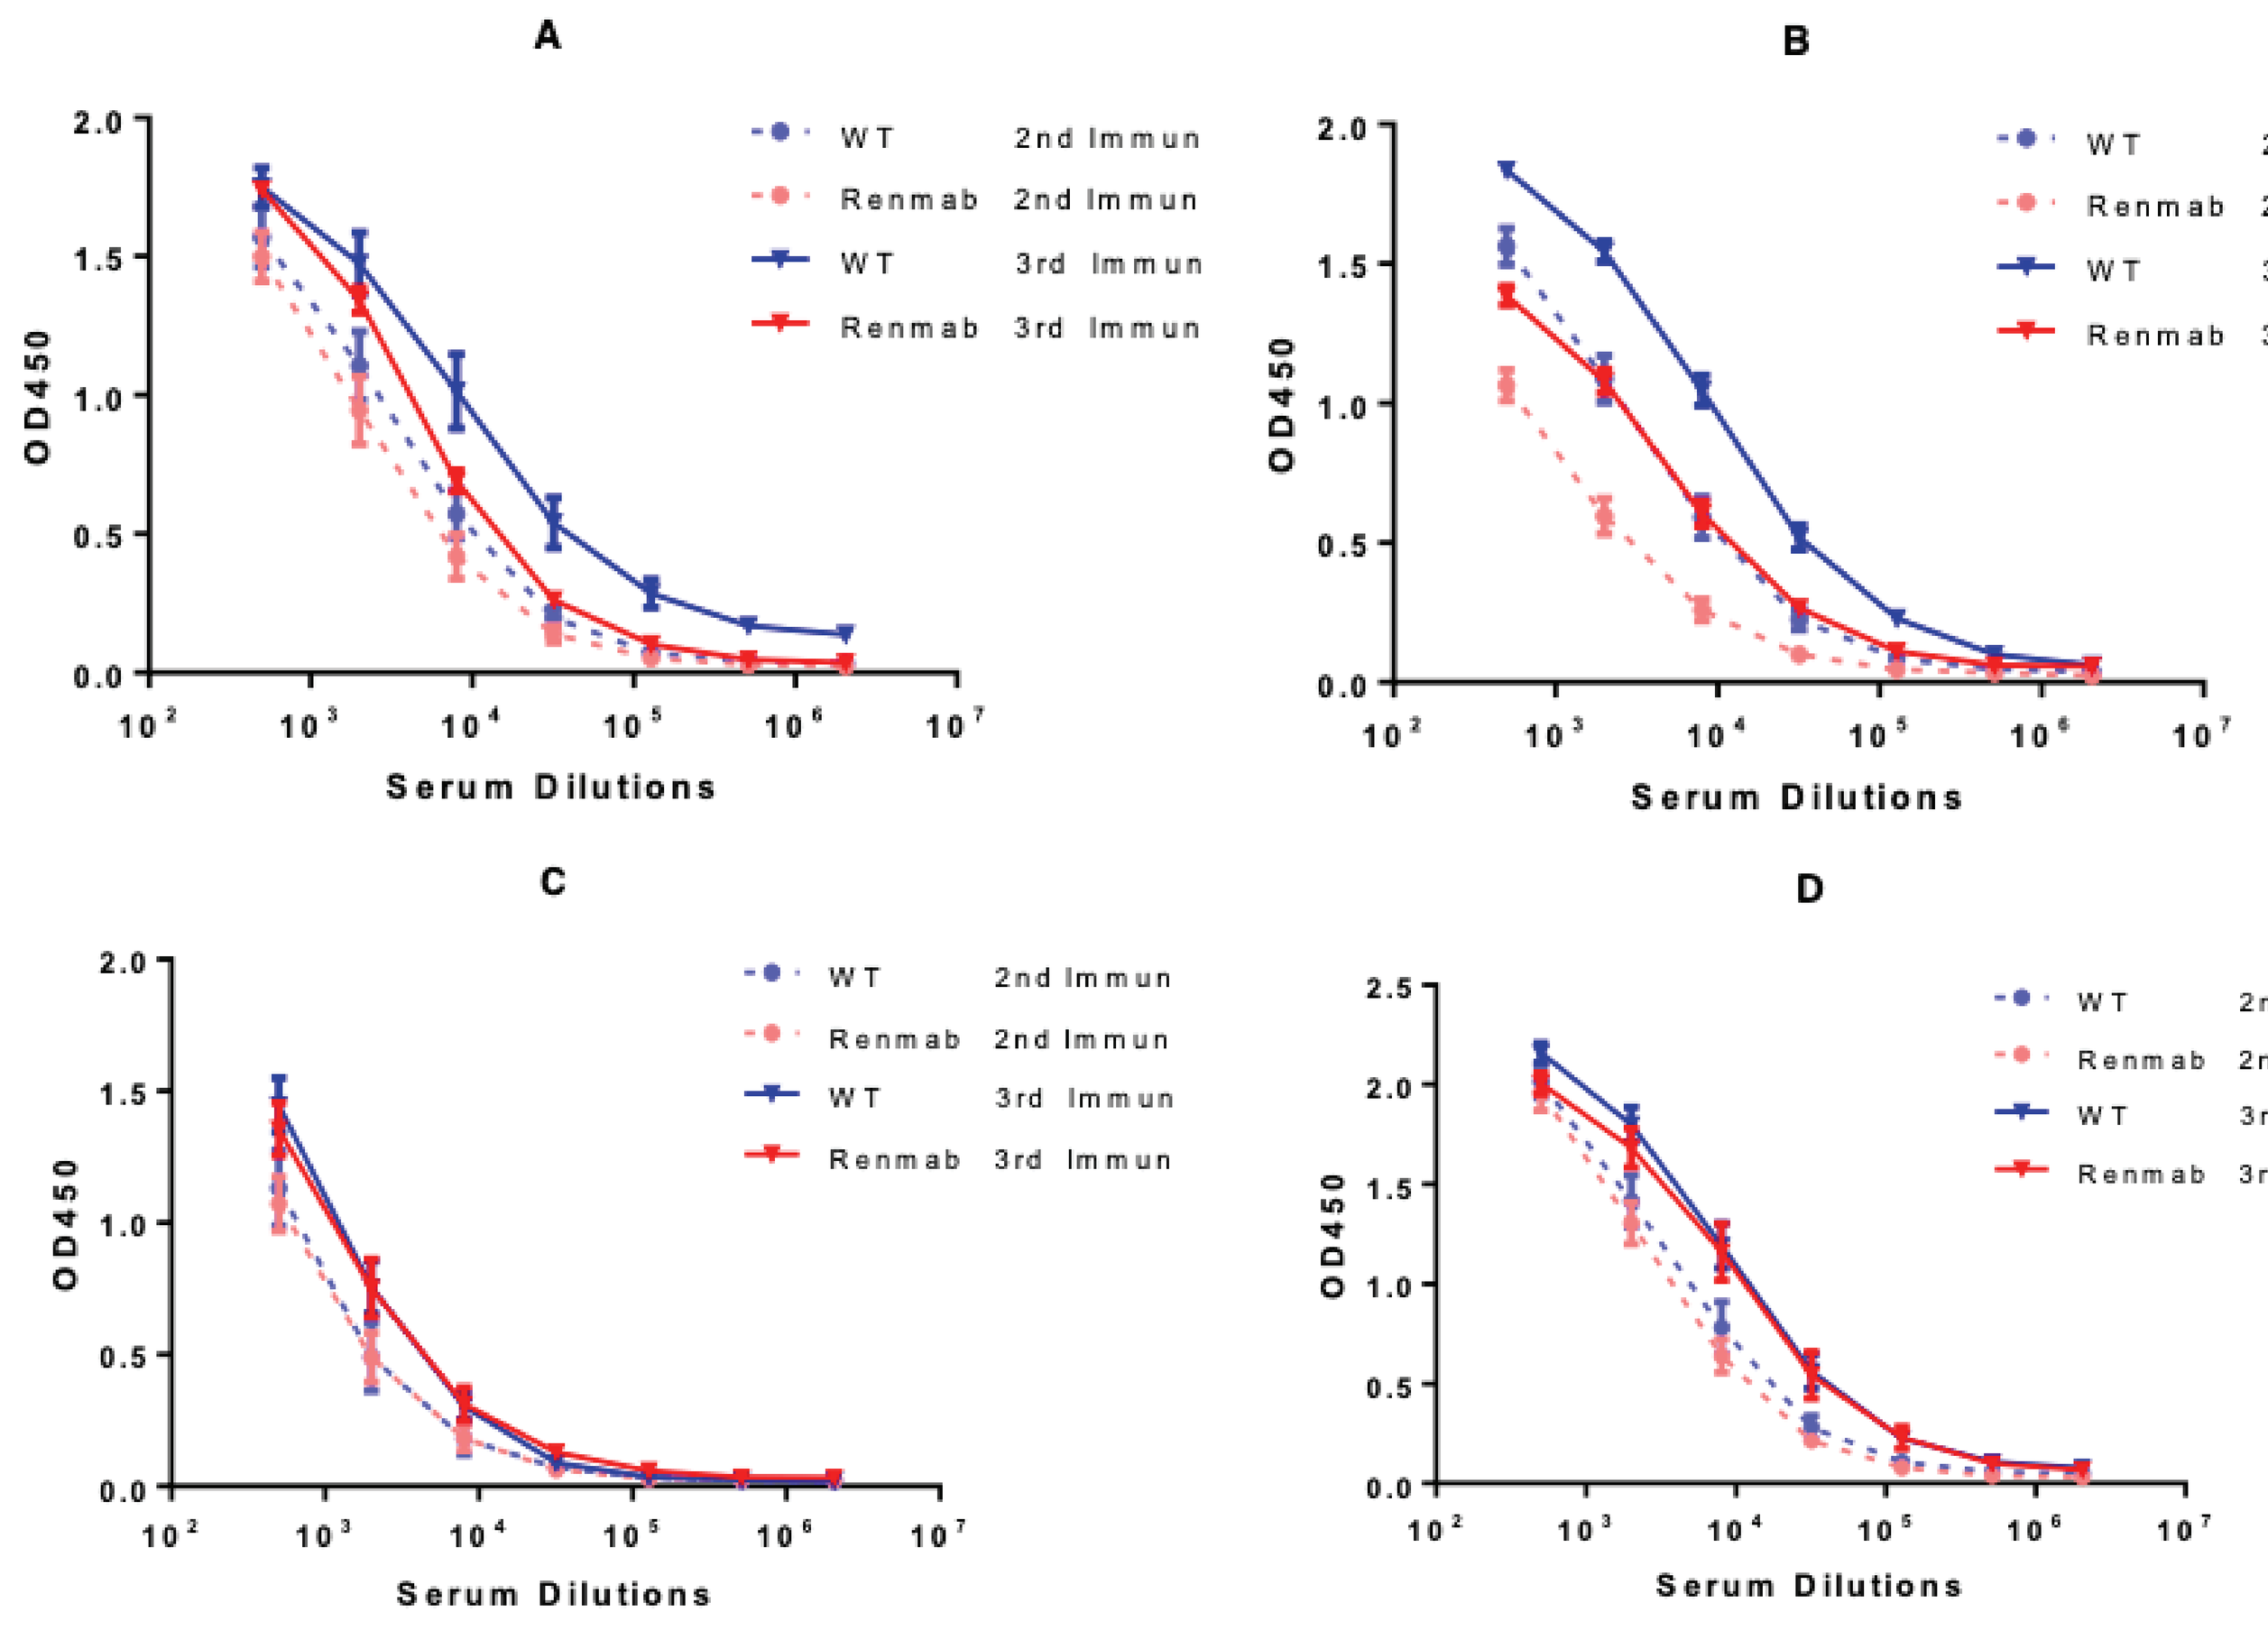
A
B
C
D
OD450
Serum Dilutions
WT 2nd Immun
Renmab 2nd Immun
WT 3rd Immun
Renmab 3rd Immun

e

RenMab™ Mice Show Normal Ig Subtypes, Suggesting Successful Class Switch

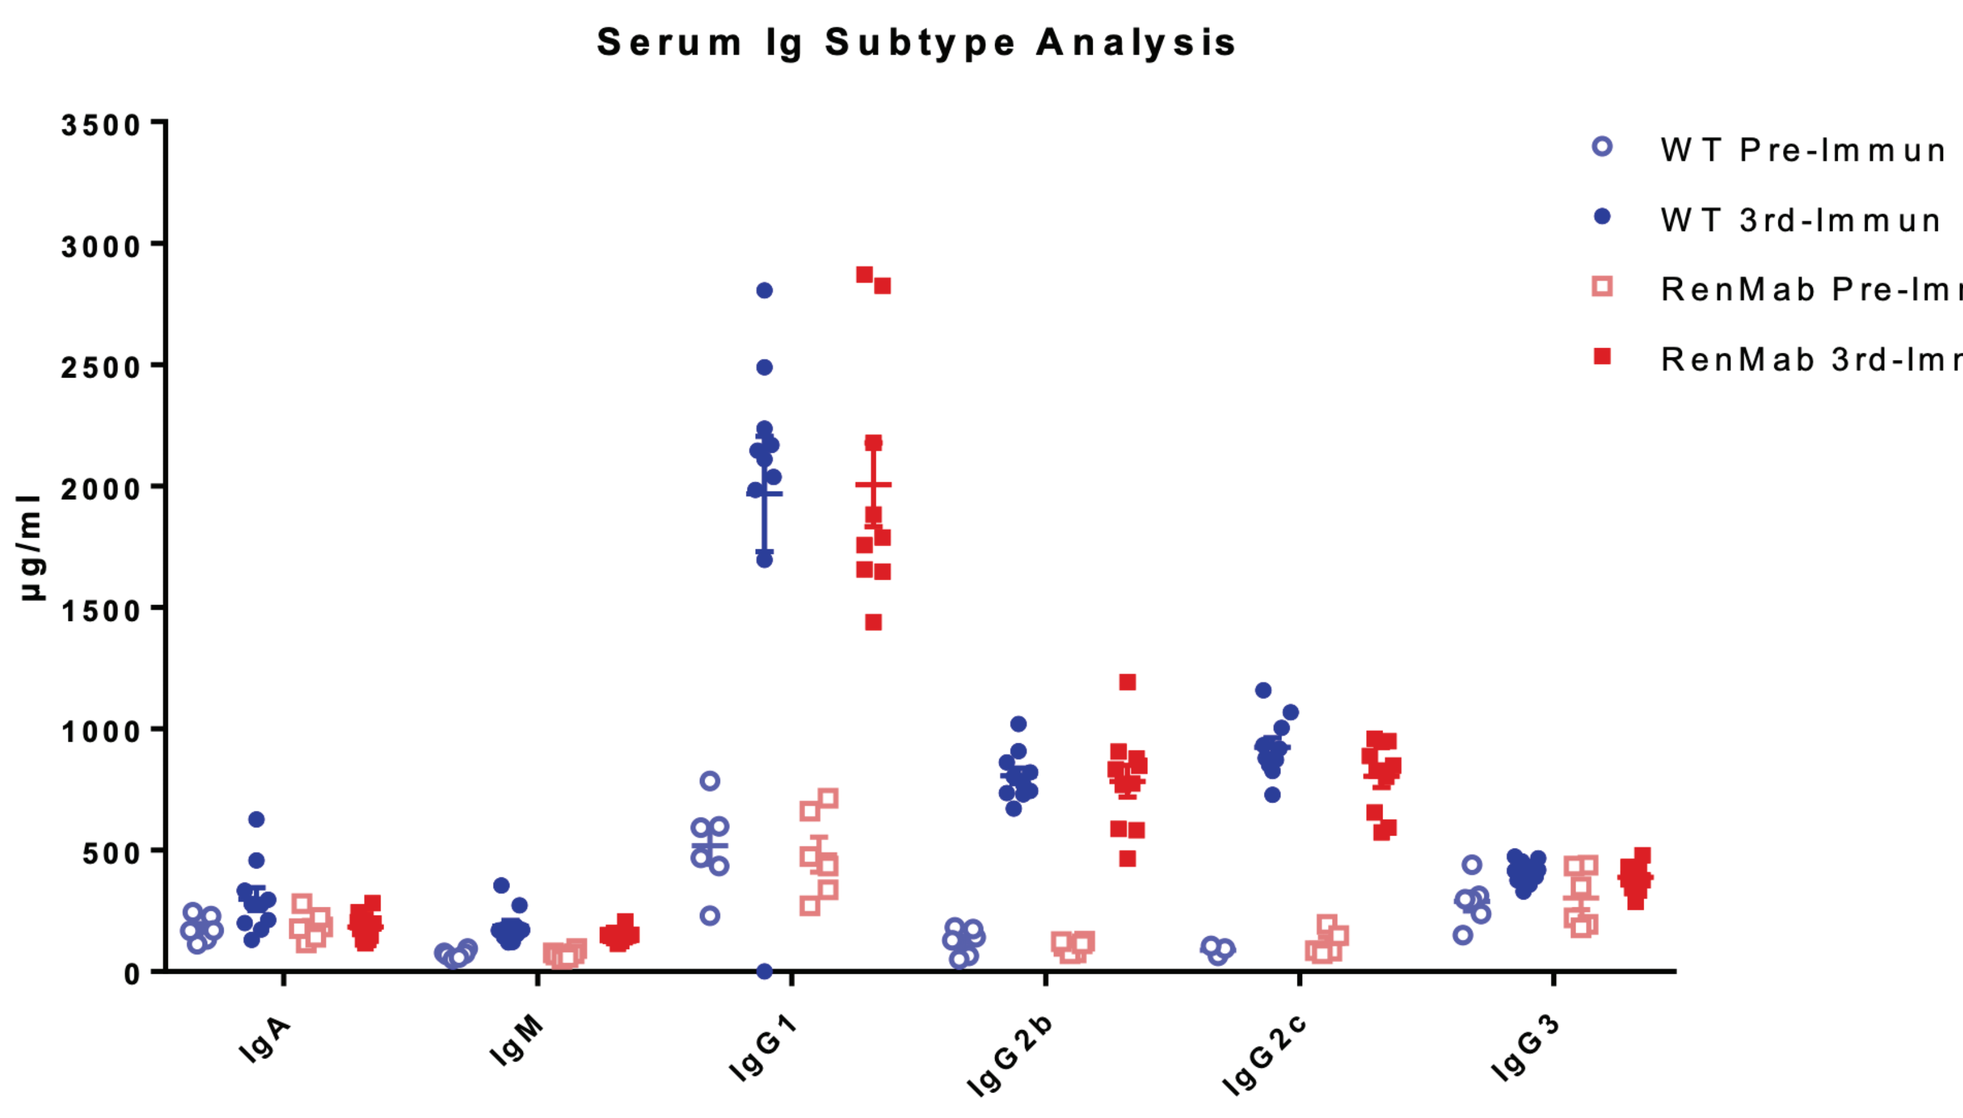
Serum Ig Subtype Analysis
μg/ml
WT Pre-Immun
WT 3rd-Immun
RenMab Pre-Immun
RenMab 3rd-Immun
IgA
IgM
IgG1
IgG2b
IgG2c
IgG3

No significant differences in serum level of IgA, IgG1, IgG2b, IgG2c, IgG3 and IgM were observed between RenMab™ and wild type mice before and after immunization.

V. RenMab™ Mice produce high affinity antibodies

RenMab™ mice generate antibodies that have nanomolar to sub nanomolar binding affinities.

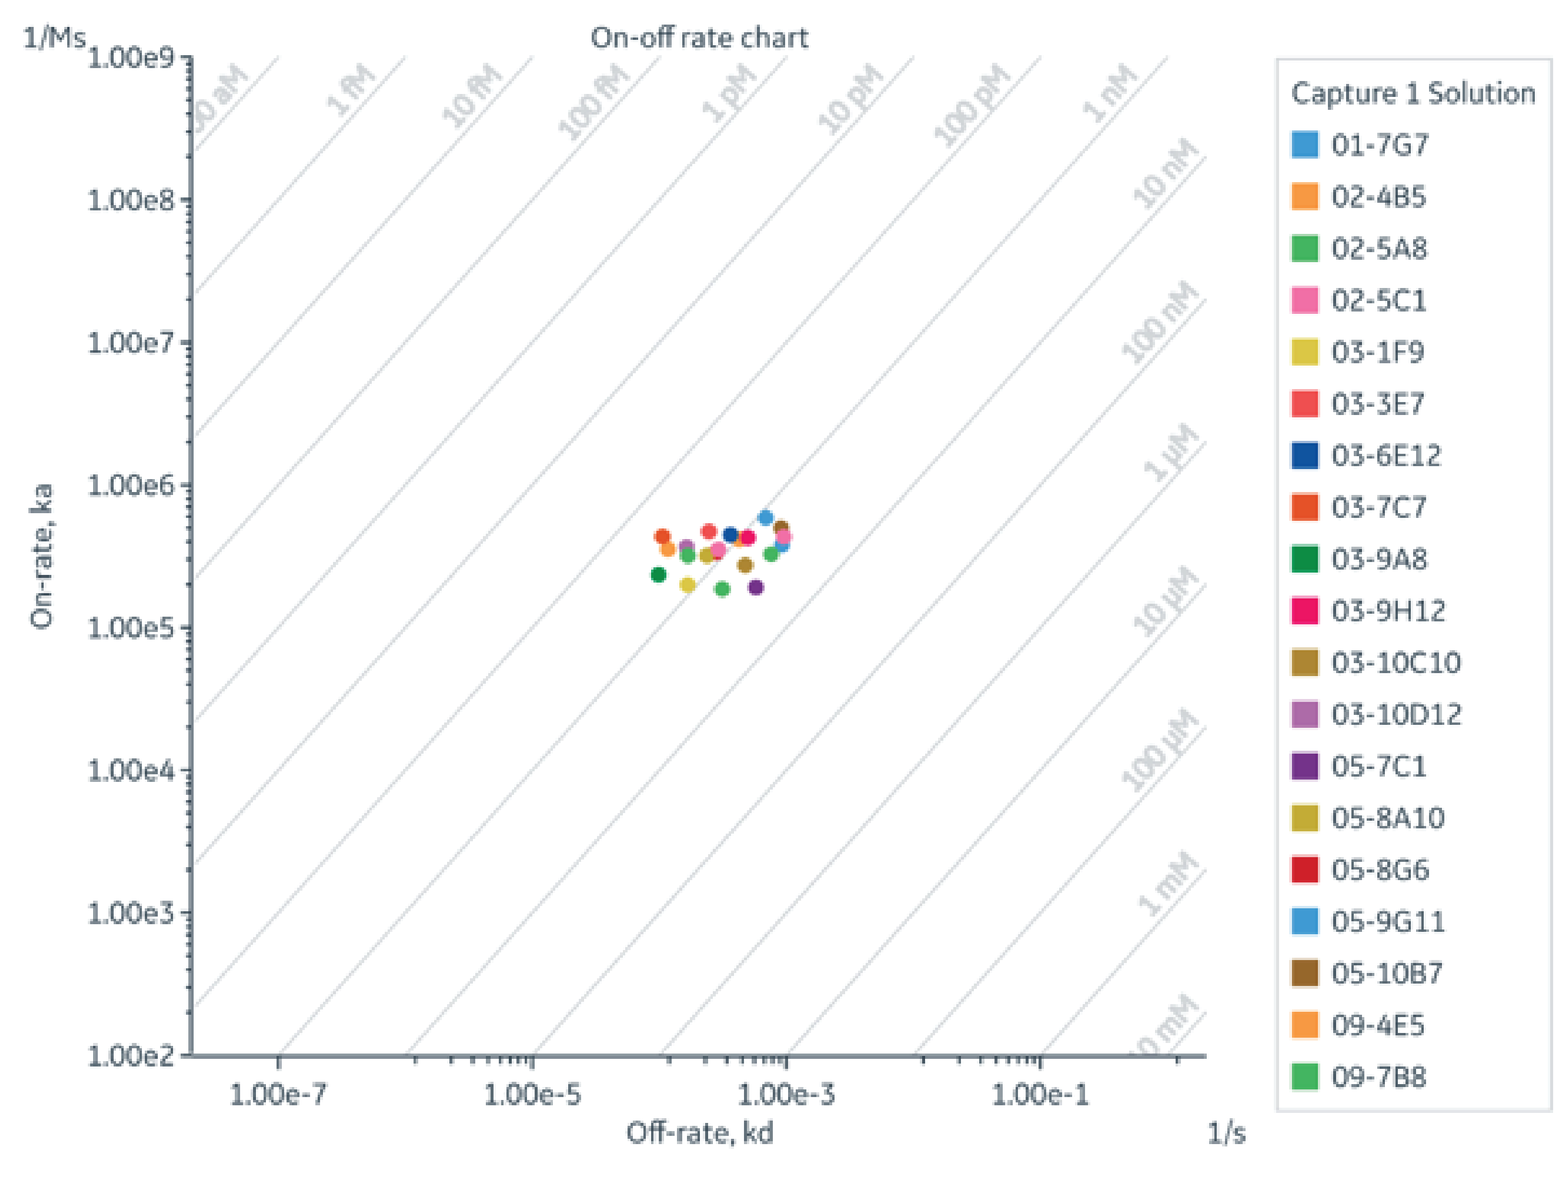
On-off rate chart
On-rate, ka
Off-rate, kd
Capture 1 Solution

Clone ID	ka (1/Ms)	kd (1/s)	KD (M)	Rmax (RU)
03-7C7	4.35E+05	1.06E-04	2.43E-10	60.1
03-10D12	3.56E+05	1.07E-04	3.00E-10	75.8
09-4E5	3.54E+05	1.17E-04	3.30E-10	98.3
03-9A8	2.35E+05	9.86E-05	4.20E-10	68.0
03-3E7	4.72E+05	2.46E-04	5.22E-10	61.8
02-5A8	3.20E+05	1.68E-04	5.24E-10	56.5
05-8A10	3.19E+05	2.38E-04	7.44E-10	68.7
03-6E12	4.47E+05	3.63E-04	8.14E-10	58.5
05-8G6	3.37E+05	2.81E-04	8.32E-10	85.9
03-1F9	2.00E+05	1.67E-04	8.35E-10	90.9
02-5C1	3.67E+05	3.63E-04	9.90E-10	34.6
02-4B5	4.16E+05	4.25E-04	1.02E-09	37.6
03-9H12	4.26E+05	4.98E-04	1.17E-09	54.5
05-9G11	5.89E+05	6.87E-04	1.17E-09	62.5
09-7B8	1.86E+05	3.13E-04	1.68E-09	24.7
03-10C10	2.74E+05	4.74E-04	1.73E-09	4.9
05-10B7	4.99E+05	9.09E-04	1.82E-09	38.6
02-5C1	4.34E+05	9.54E-04	2.20E-09	31.9
02-5A8	3.27E+05	7.59E-04	2.32E-09	30.3
01-7G7	3.80E+05	9.23E-04	2.43E-09	68.3
05-7C1	1.91E+05	5.75E-04	3.01E-09	55.6

CONCLUSIONS

In conclusion, RenMab™ Mouse platform provides:

• Fully human antibody repertoire diversity of heavy chain and kappa light chain.
• Conservation of regulatory elements of both human (within VDJ loci) and mouse (outside VDJ loci).
• Normal B cell development and immune cell profile.
• Robust immune response comparable to wild type mouse.
• Antibody affinity of nanomolar to sub nanomolar range.
• RenMab™-KO mouse for challenging targets such as proteins with high homology between human and mouse or GPCR/Ion channels.
• Recent addition of RenLite ™ mice with humanized heavy chain and fixed human light chain, suitable for bispecific antibody discovery

Information available at www.renmab.com or send inquiry to: info@biocytogen.com.

Combined with existing inventory of single, double or triple target humanized mouse models and a world-leading proprietary gene editing platform, Biocytogen is dedicated to providing full support on every stage of your antibody discovery journey.

www.biocytogen.com | info@biocytogen.com